Feasibility of Subcutaneous Entecavir Implants for Chronic Hepatitis B Treatment

Steven J. Henry; Stephanie E. Barrett; Seth Forster; Ryan S. Teller; Zhen Yang; Gregory J. Doto; Michael P. Ruth; Takayuki Tsuchiya; Lee J. Klein; Marian E. Gindy

Merck Research Laboratories (West Point, PA), Merck & Co., Inc., Kenilworth, NJ, USA

MERCK

Abstract

Patient compliance in the treatment of chronic infectious diseases is critical in preventing disease progression but often hampered by the burden of a lifelong pill regimen.[1] Entecavir (ETV) is a first-line antiretroviral agent for the treatment of chronic hepatitis B administered as a once-daily oral pill or solution.[2] The aim of our work was to evaluate the feasibility of long-acting implantable formulations of ETV to reduce the compliance burden on patients. To this end, we engineered hot melt extrudates and coated tablets for sustained ETV release in the subcutaneous (SC) space.

A variety of pharmacokinetic release profiles were achieved in rats by tuning the rate-controlling properties within a given modality. While 6-month SC delivery of ETV was demonstrated, local inflammation and necrotic tissue were observed proximal to the implant. Although SC ETV was not well tolerated at high input rates, the demonstrated implant longevity was a substantial increase over previous efforts reported in the literature.[3] The modalities employed herein may have applicability to other therapeutic agents for long-acting hepatitis B treatment.

A Problem of Global Proportions: 240-360 Million Individuals With CHB[2,4]

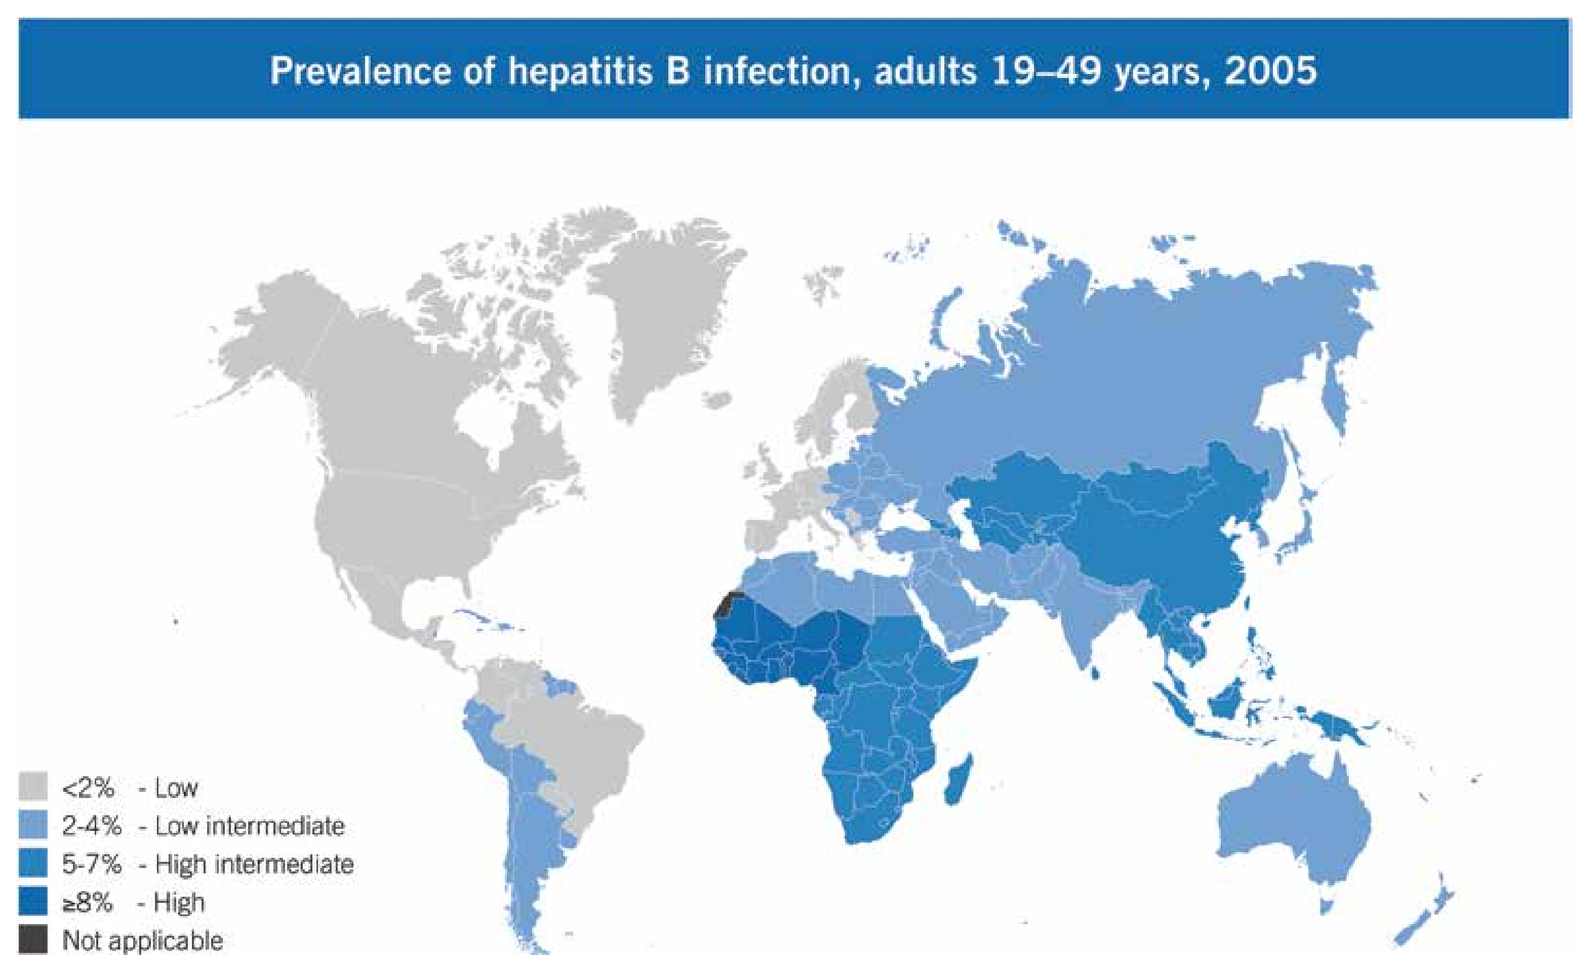
Prevalence of hepatitis B infection, adults 19–49 years, 2005
<2% - Low
2-4% - Low intermediate
5-7% - High intermediate
≥8% - High
Not applicable

Entecavir (ETV) is an off-patent first-line nucleotide reverse transcriptase inhibitor taken at 0.5 mg/day orally for a patient's lifetime. The molecule is shown in its intracellular triphosphorylated form to the right.

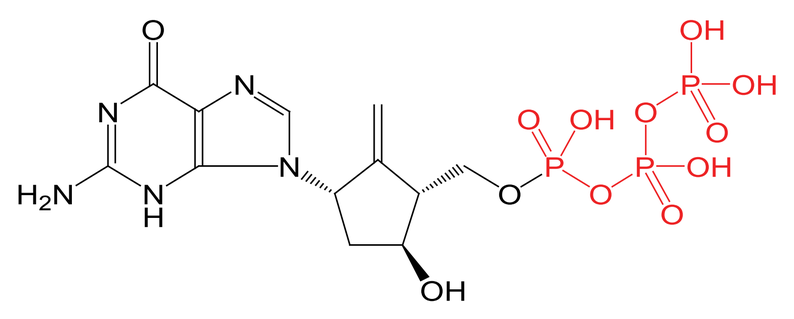

Hot Melt Extrudates and Coated Tablets

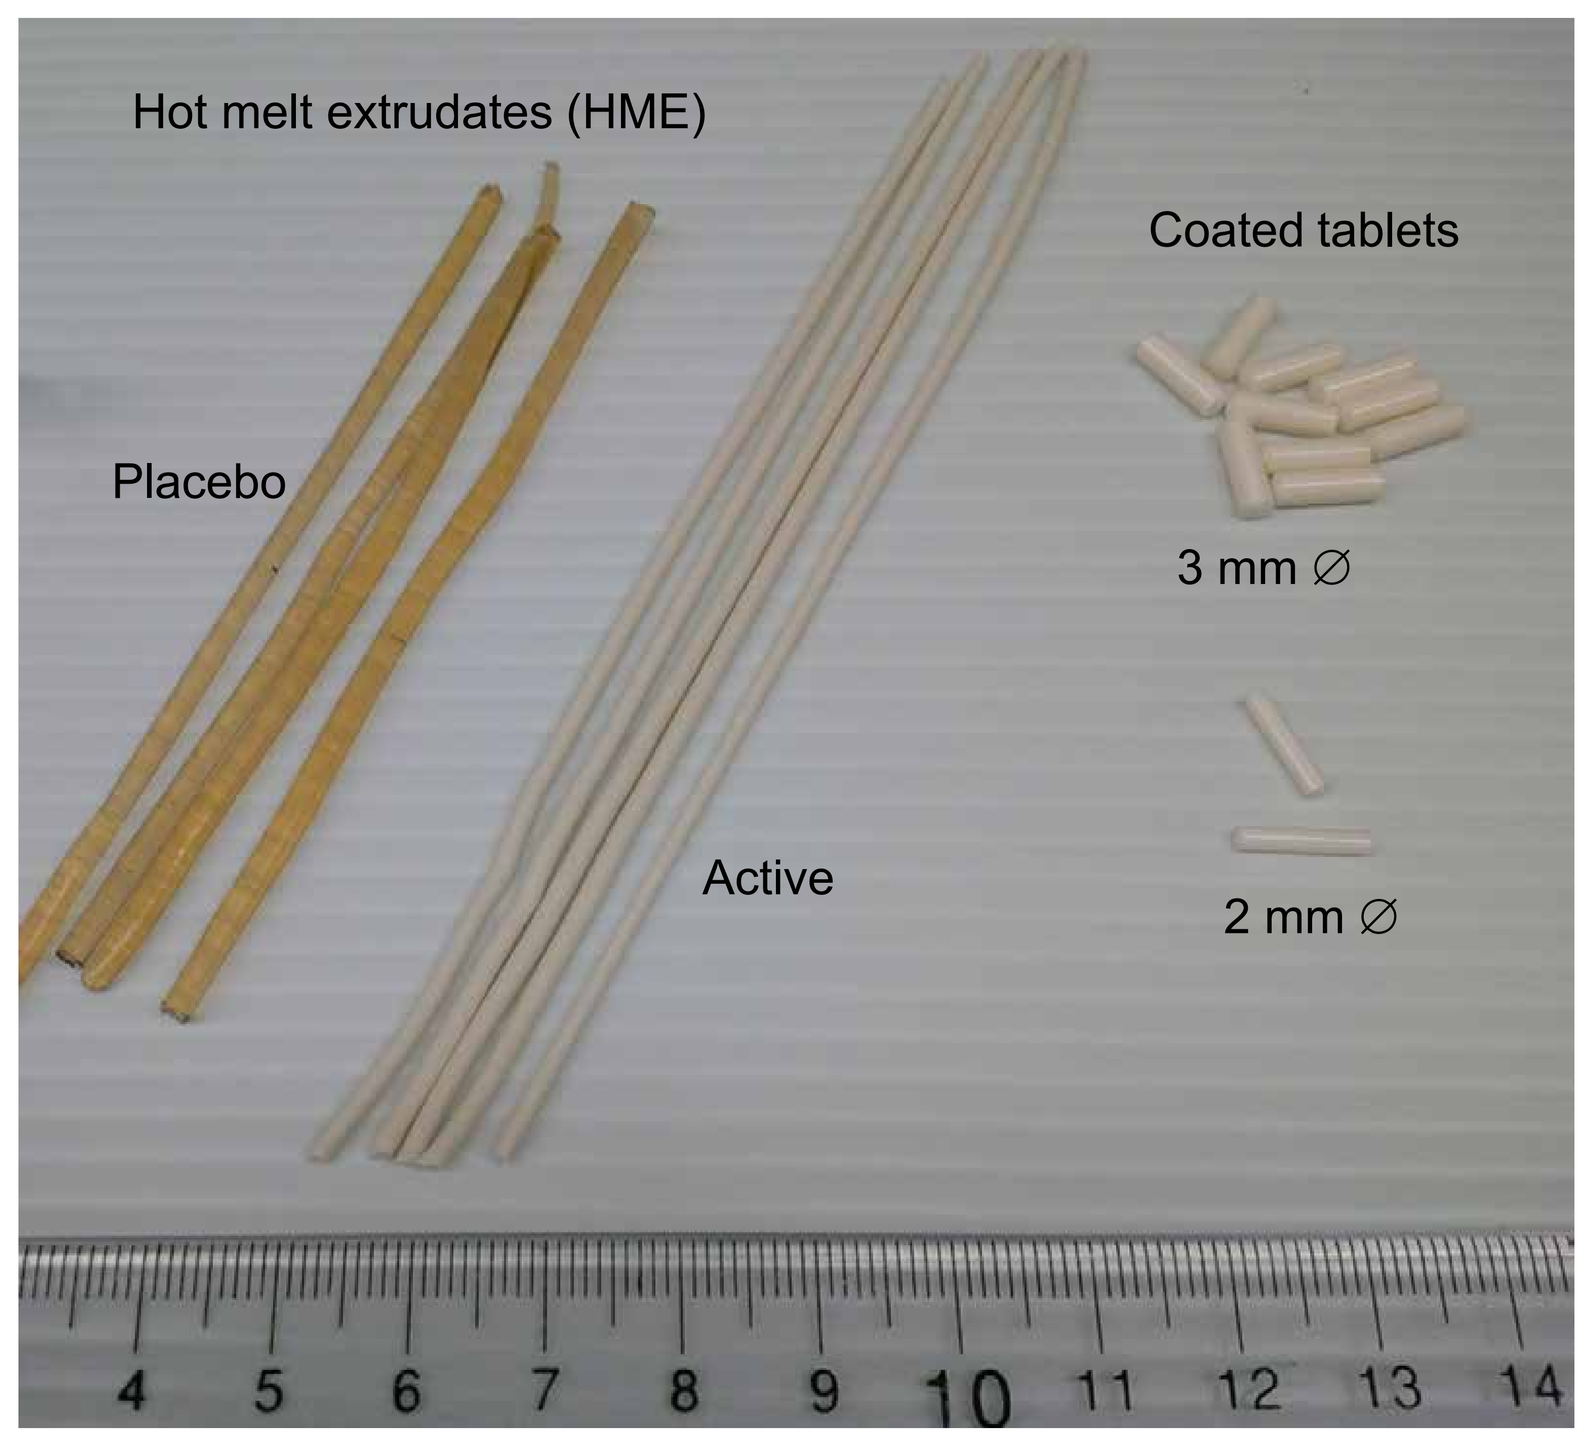
Hot melt extrudates (HME)
Coated tablets
Placebo
3 mm ∅
Active
2 mm ∅

Polymers Evaluated

Hot melt extrudates (HME)	Polymer coated tablets (Tab)
1X DL poly(D,L-lactide) (PLA)	2.3X DL uncoated (bare)
1X DL polycaprolactone (PCL)	2.3X DL polyurethane† (PU)
	2.3X DL poly(D,L-lactide-co-glycolide) (PLGA)
	2.3X DL poly(D,L-lactide-co-caprolactone) (PLCL)

†All polymers evaluated were bioerodible with exception of PU.

References

1. Buell KG, et al. *AIDS Care*. 2016;28(2):242-246. DOI: 10.1080/09540121.2015.1074653
2. World Health Organization. Guidelines for … chronic hepatitis B infection. 2015. ISBN: 9789241549059
3. Lim JL, et al. *Int J Pharm*. 2015;490(1-2):265-272. DOI: 10.1016/j.ijpharm.2015.05.049
4. Shepard CW, et al. *Epidemiol Rev*. 2006;28:112-125. DOI: 10.1093/epirev/mxj009

Tuning HME *In Vitro* Release

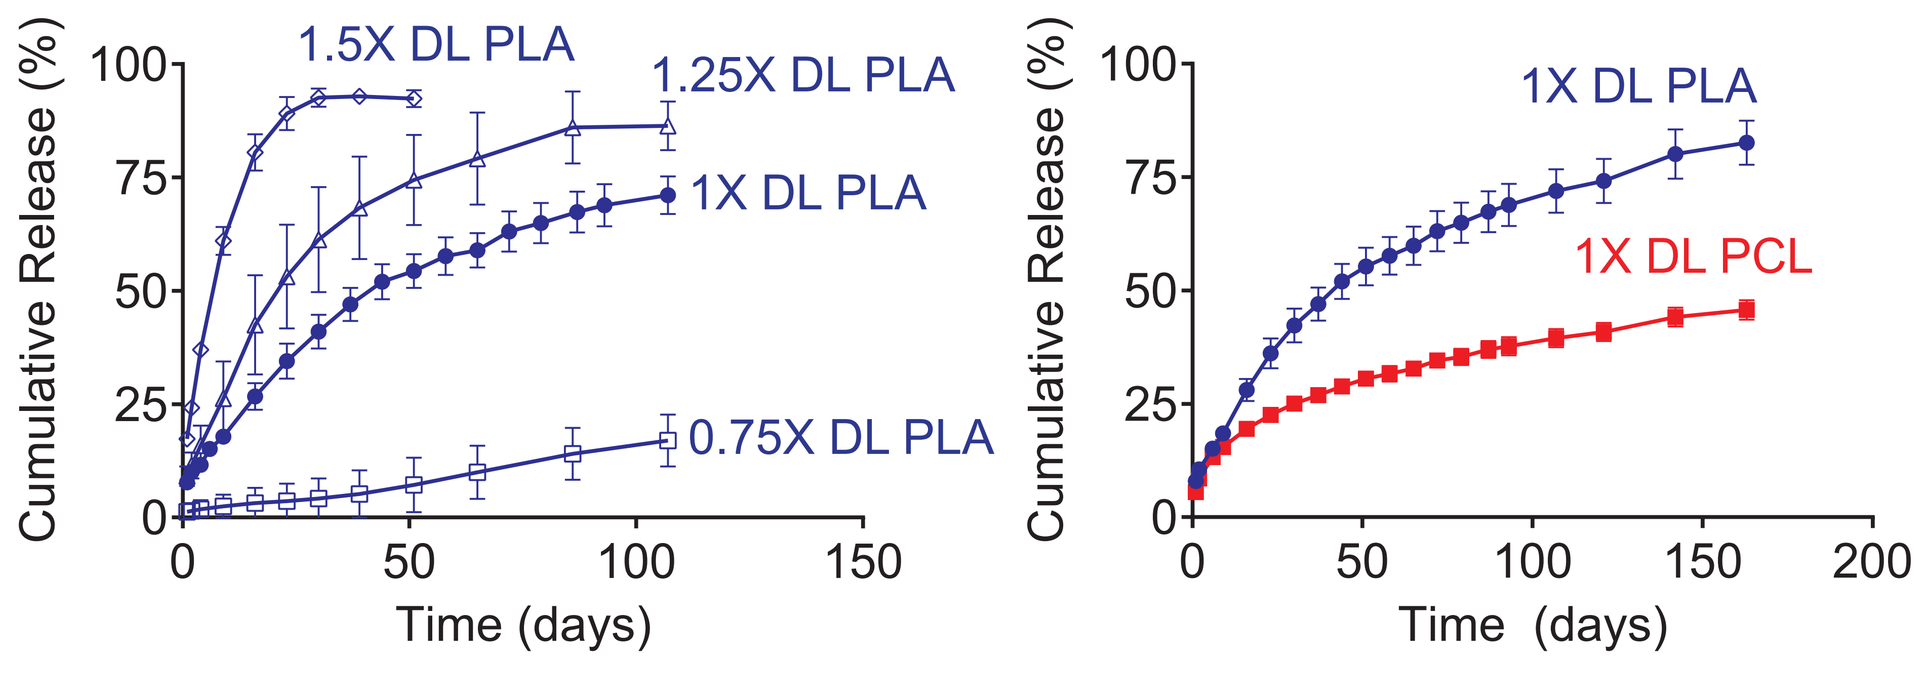
Cumulative Release (%)
1.5X DL PLA
1.25X DL PLA
1X DL PLA
0.75X DL PLA
1X DL PCL
Time (days)

Error bars = ± SD (n = 3 replicates per condition)

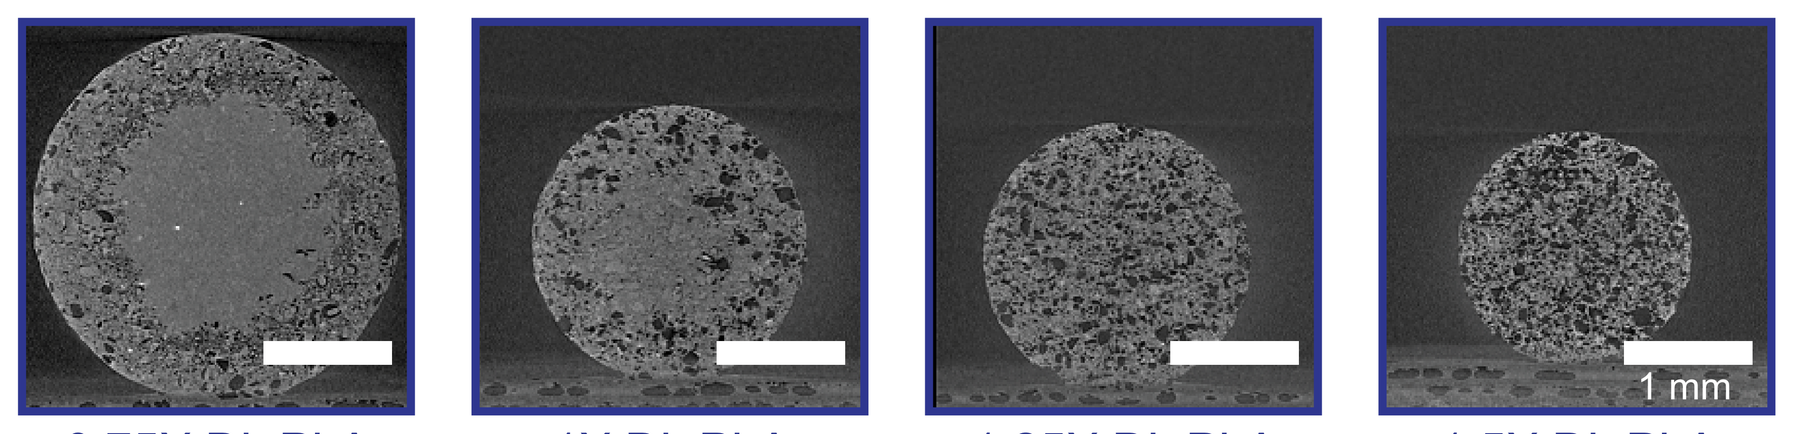
1 mm

0.75X DL PLA Day 107 | 1X DL PLA Day 107 | 1.25X DL PLA Day 107 | 1.5X DL PLA Day 51

Manual Tablet Coating: Dip-Flip-Dip

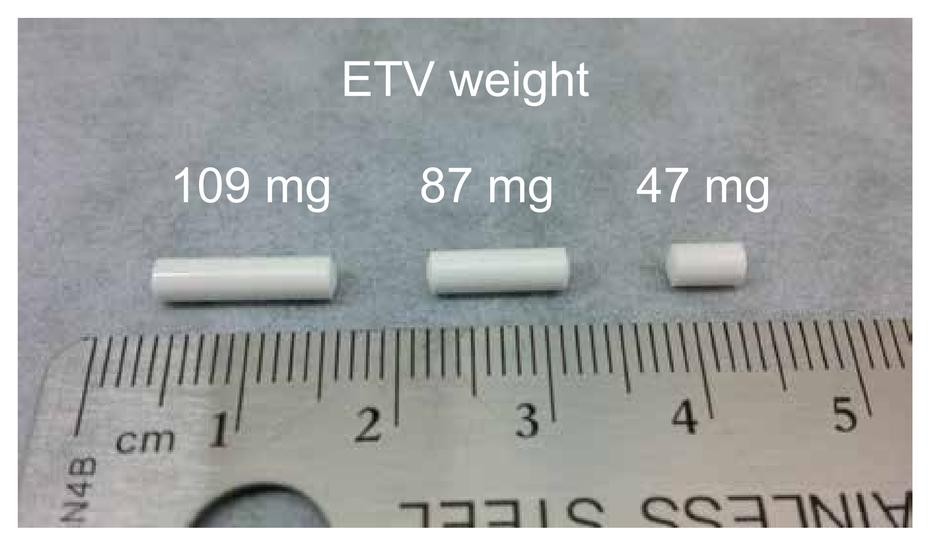
ETV weight
109 mg
87 mg
47 mg

Intrinsic ETV compressibility enables high drug load (DL) tablets by direct compression.

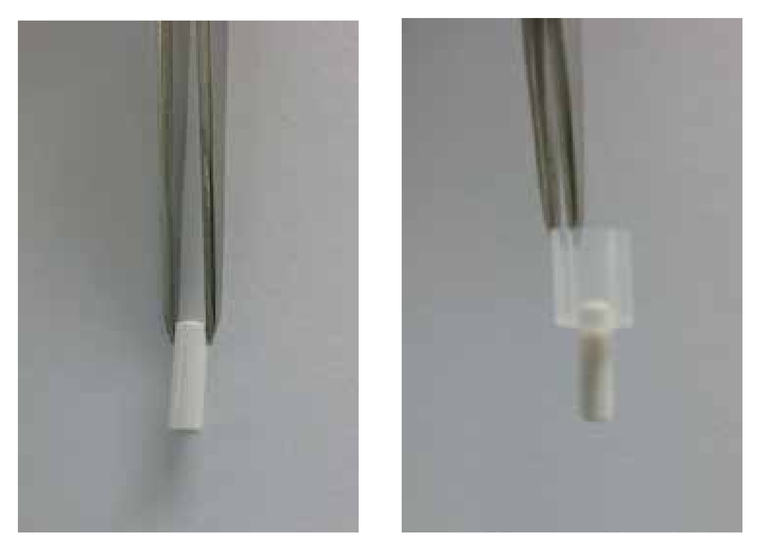
Direct gripping
Sheathed gripping

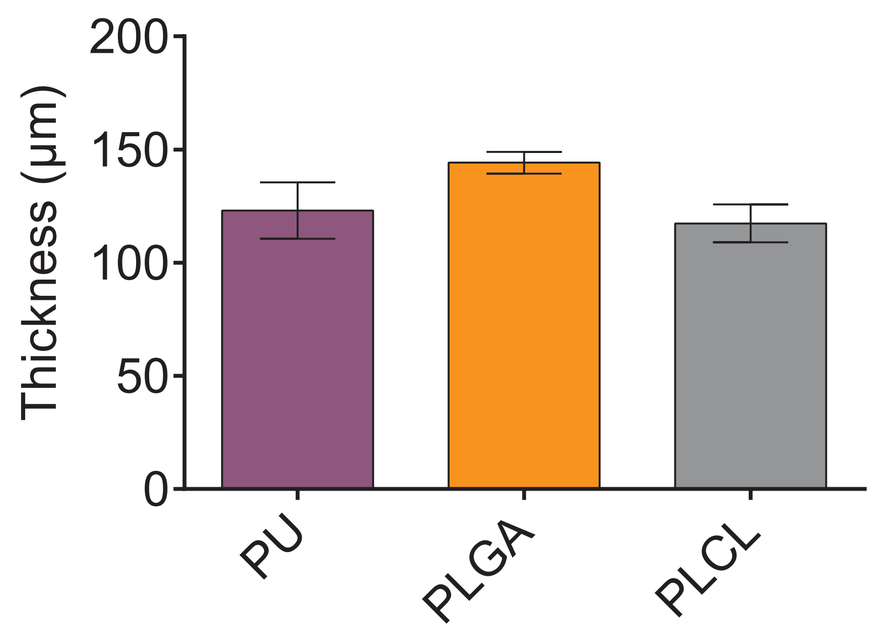
Thickness (µm)
PU
PLGA
PLCL

Error bars = ± SD (n = 3 replicates per condition)

Drying rack

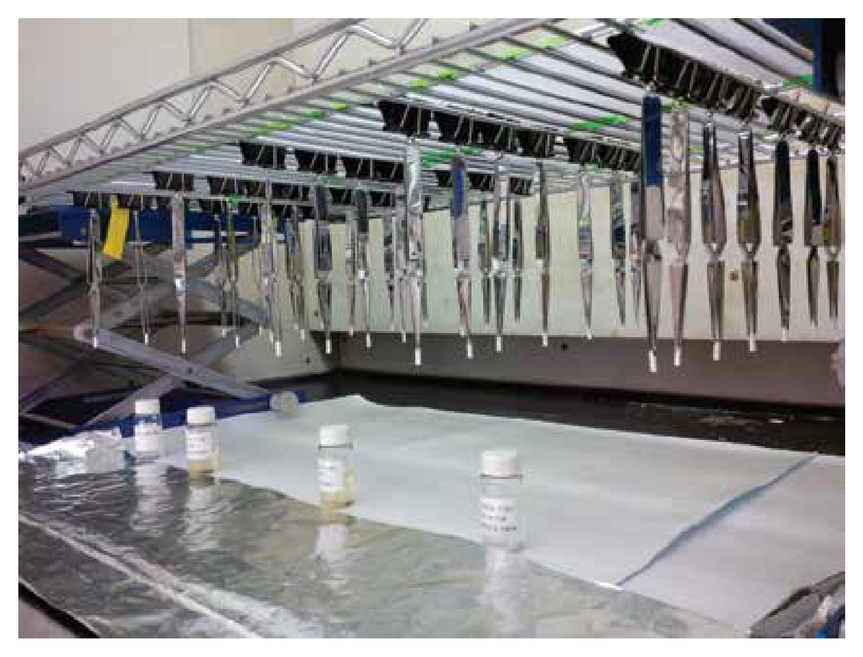

Tuning Coated Tablet *In Vitro* Release

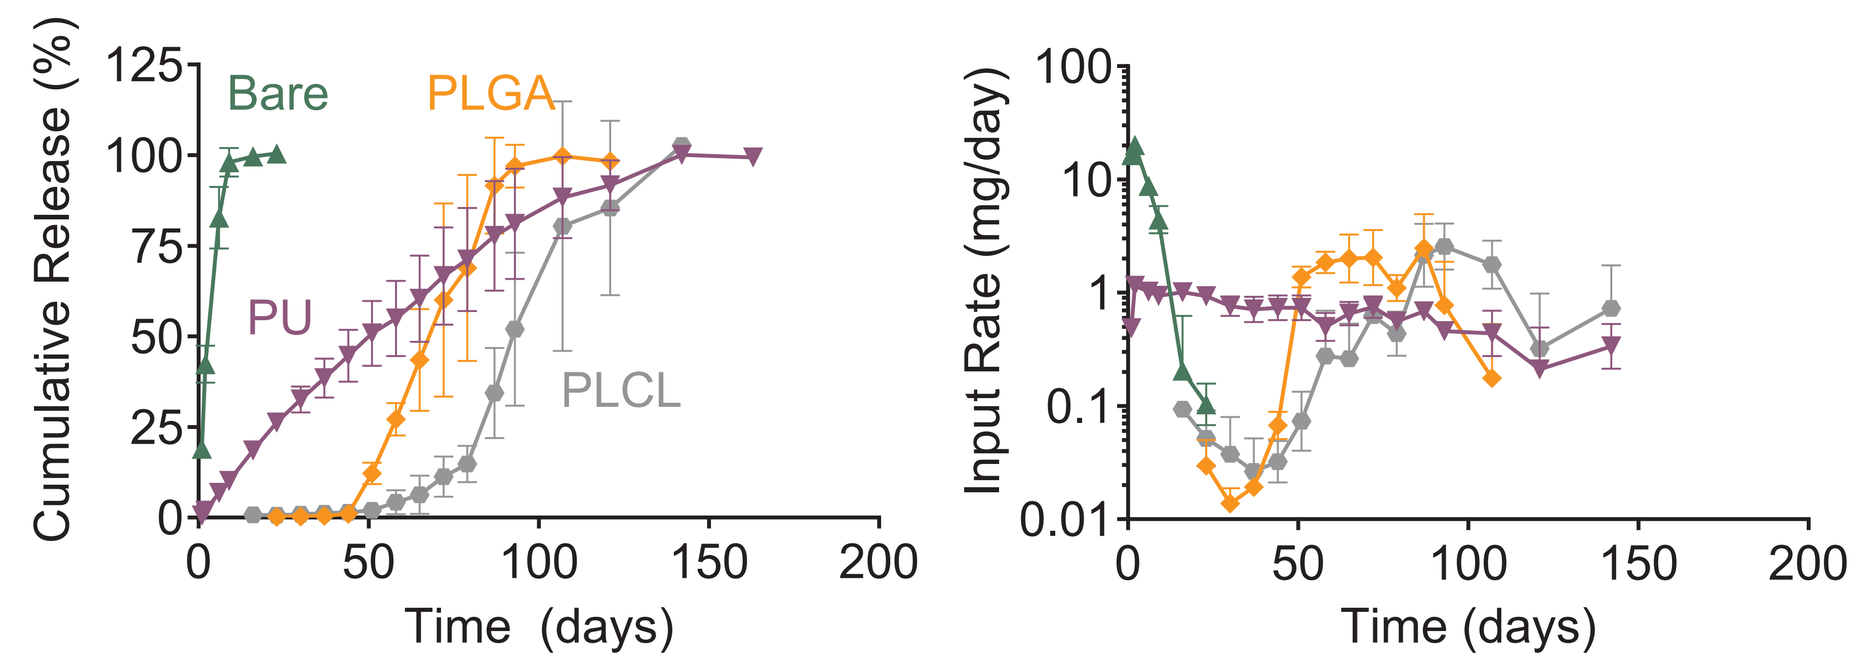
Cumulative Release (%)
Bare
PLGA
PU
PLCL
Input Rate (mg/day)
Time (days)

Error bars = ± SD (n = 3 replicates per condition)

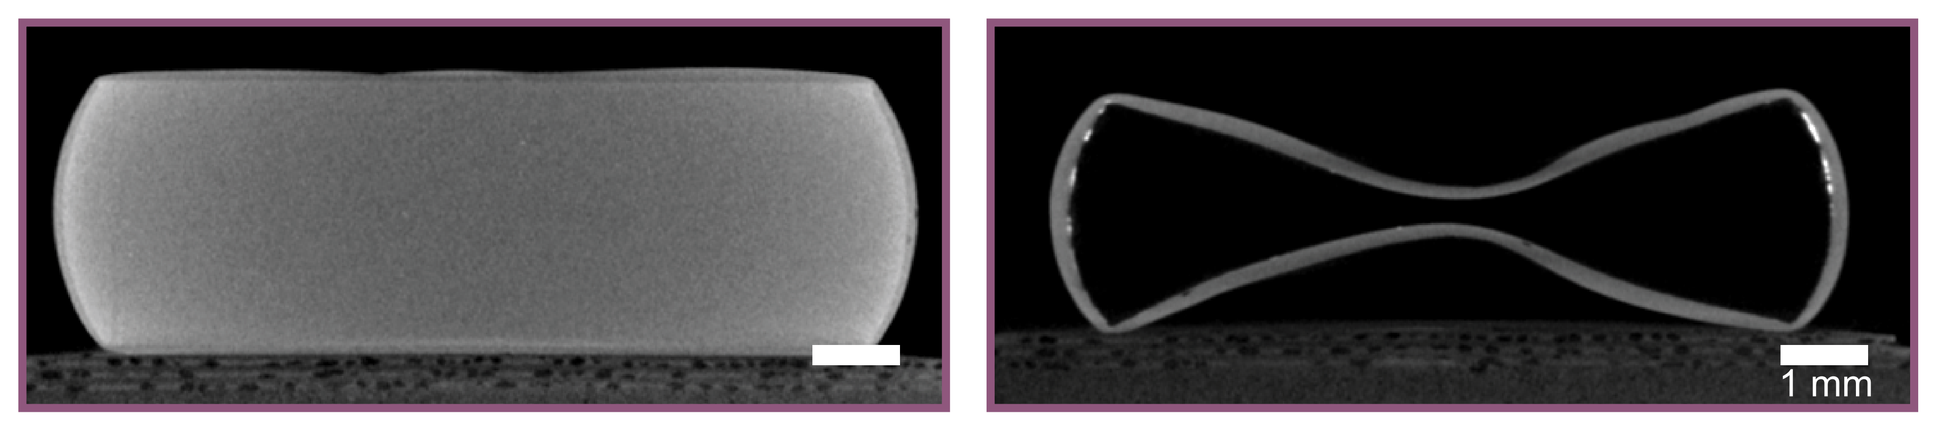
1 mm

PU Tab, Day 0 | PU Tab, Day 163

Defect-Enabled Release From PCL Tablet Suggests Utility of a Fixed-Orifice Design

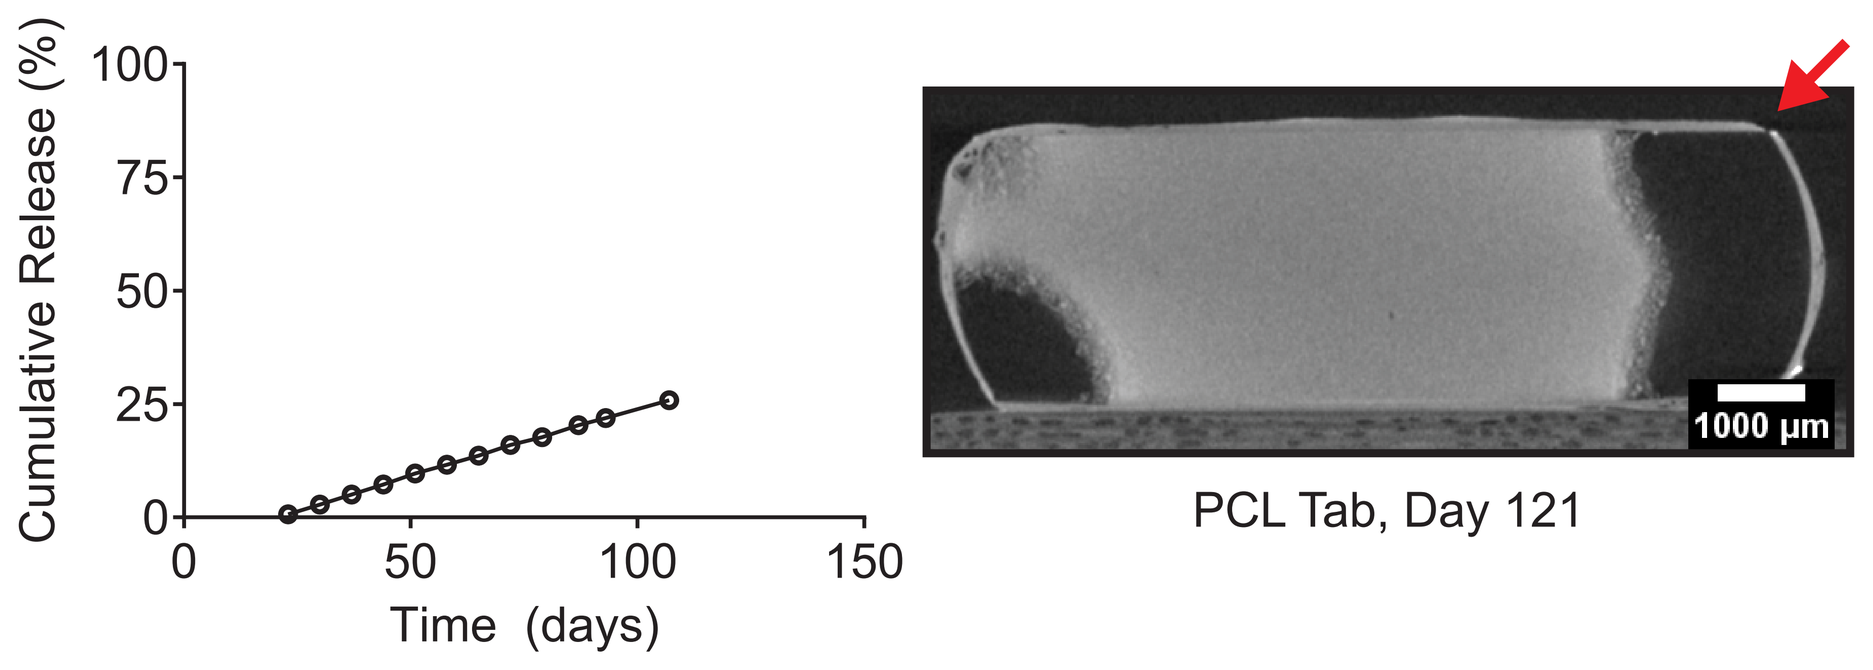
Cumulative Release (%)
Time (days)
1000 µm

PCL Tab, Day 121

1/6 replicates: Defect-enabled release (see XRCT cross section above)
5/6 replicates: No detectable release through 121 days

The defect-enabled release of ETV from PCL-coated tablets suggests that the predominate mechanism of ETV escape from PCL HMEs is through pores and channels, not via diffusion through the polymer matrix itself.

A Variety of ETV PK Profiles Were Achieved in Rats Using HMEs and Coated Tablets

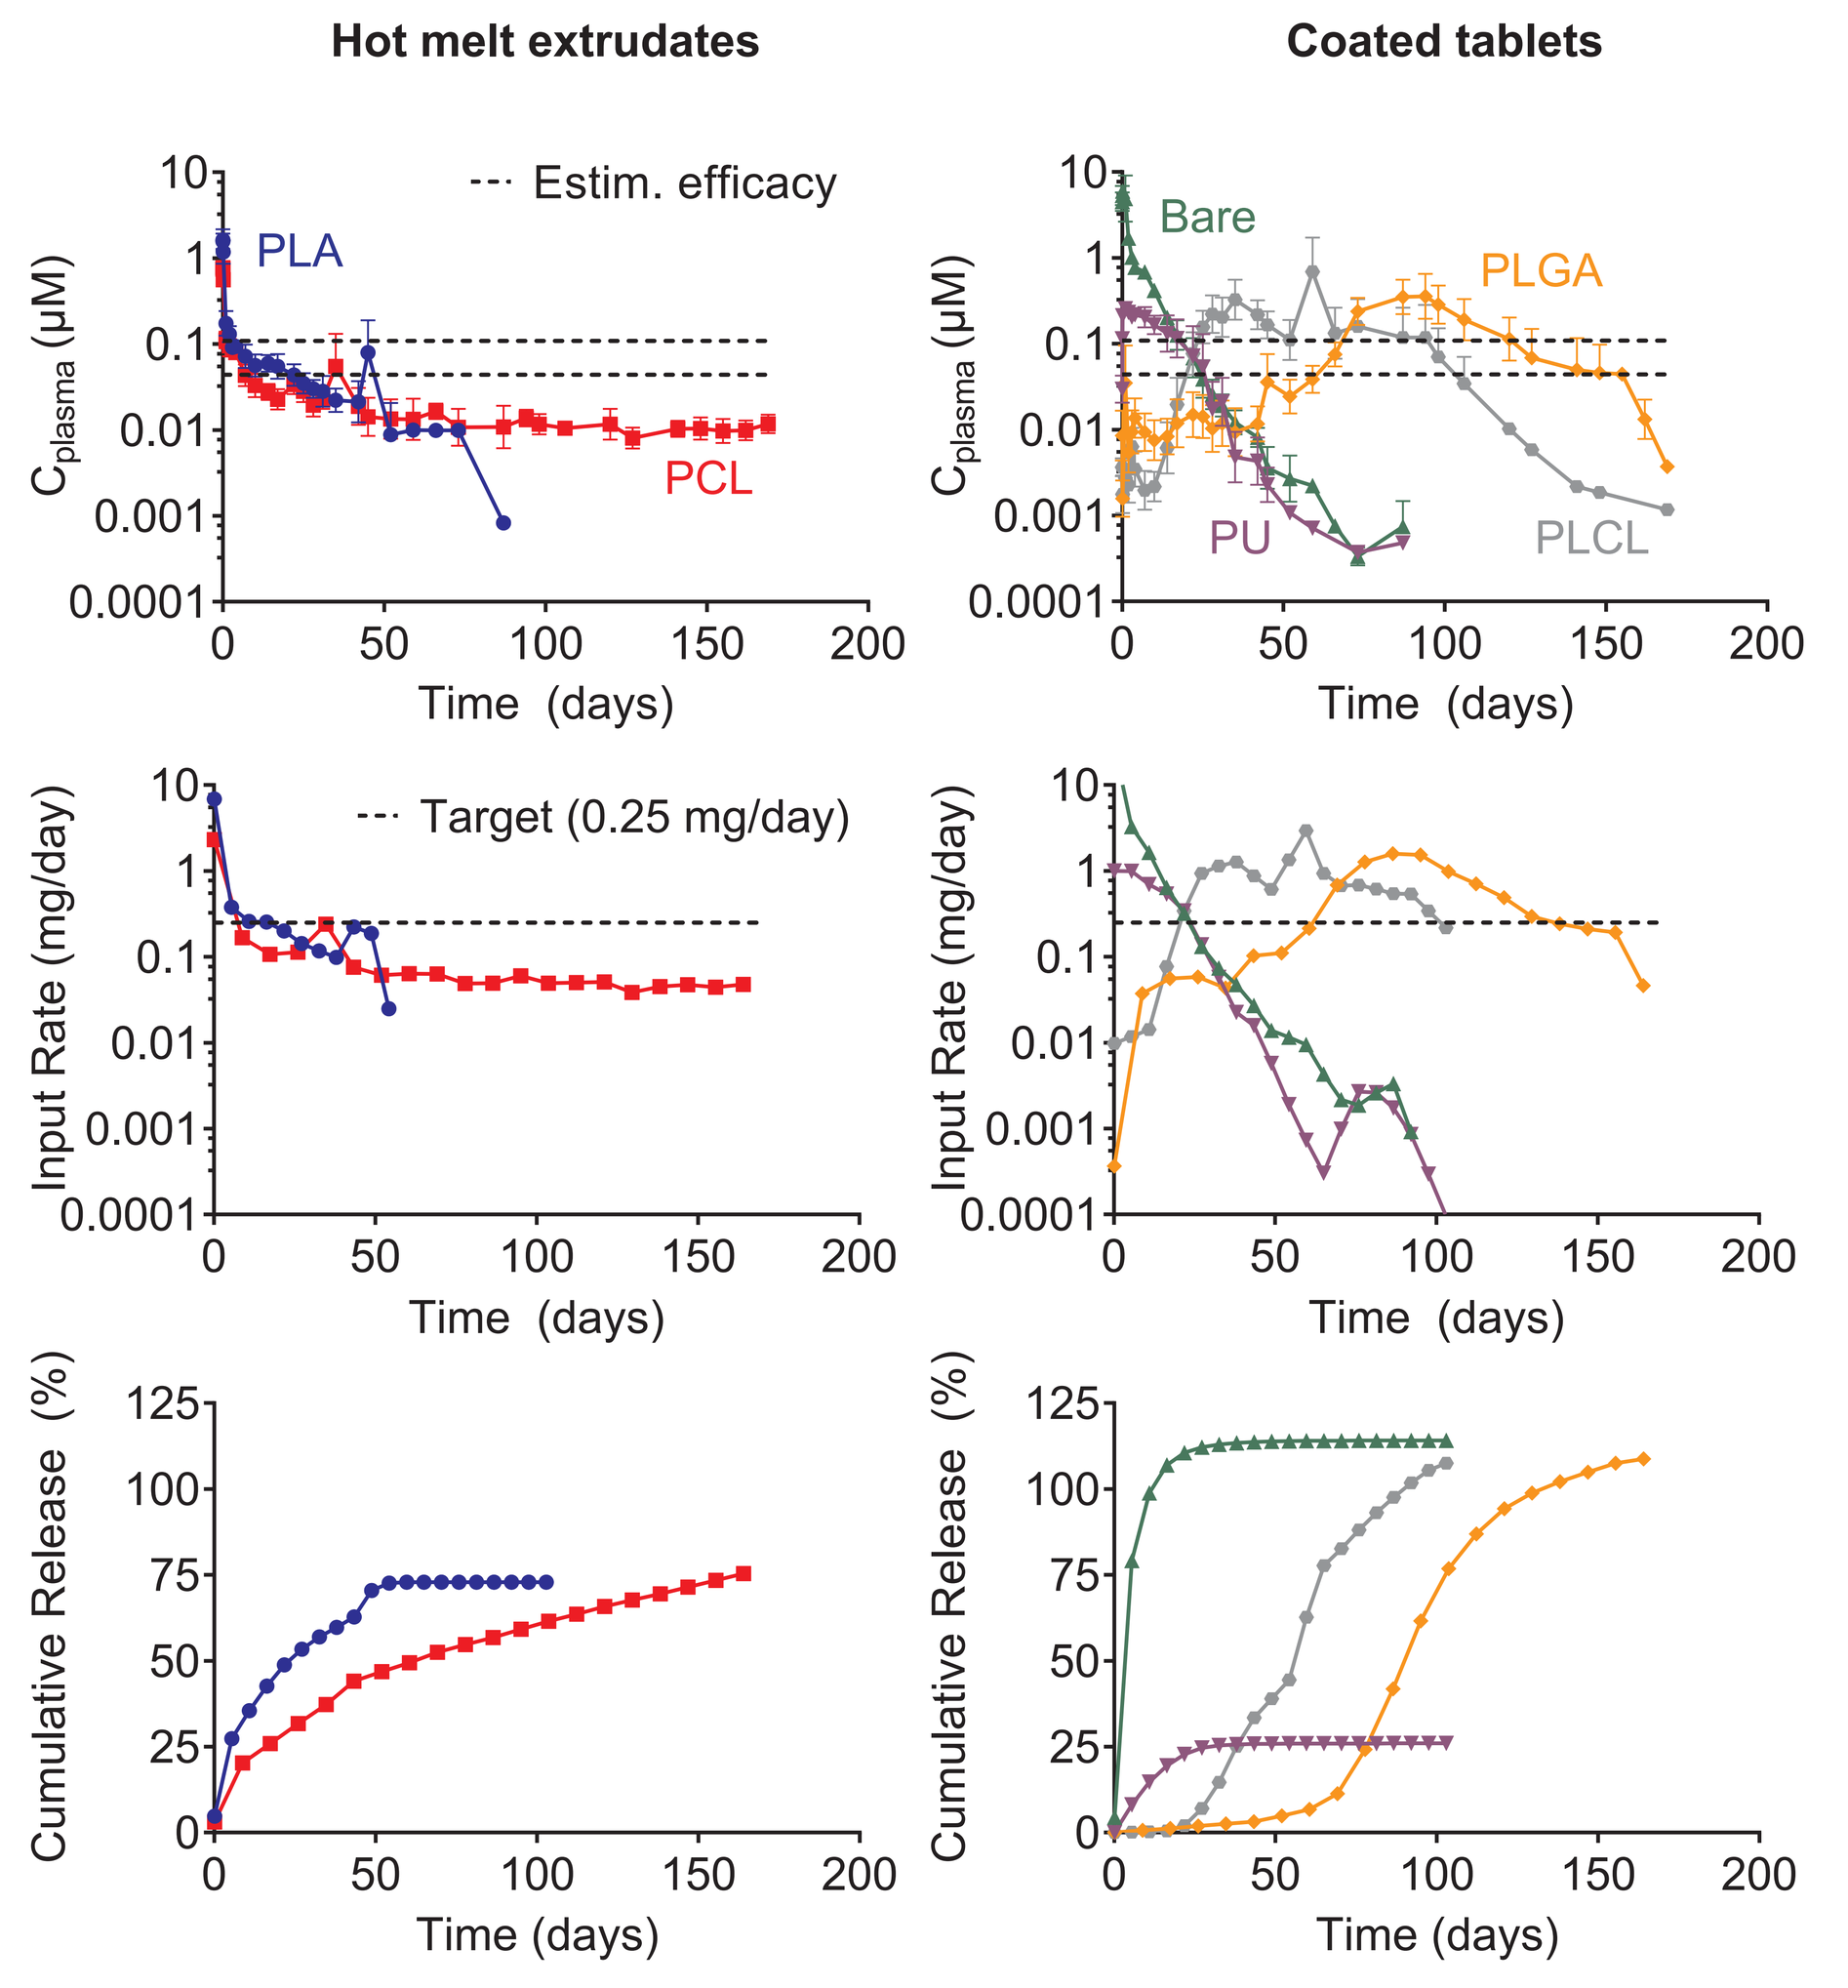
Hot melt extrudates
Coated tablets
Estim. efficacy
PLA
PCL
Bare
PLGA
PU
PLCL
Target (0.25 mg/day)
Input Rate (mg/day)
Cumulative Release (%)
Time (days)

Error bars = ± SD (n = 2-4 rats per condition)
Note: Input rate and cumulative release were result of deconvolution analysis using measured clearance rate of ETV from rats.

Chronic Subcutaneous ETV Exposure-Induced Local Inflammation

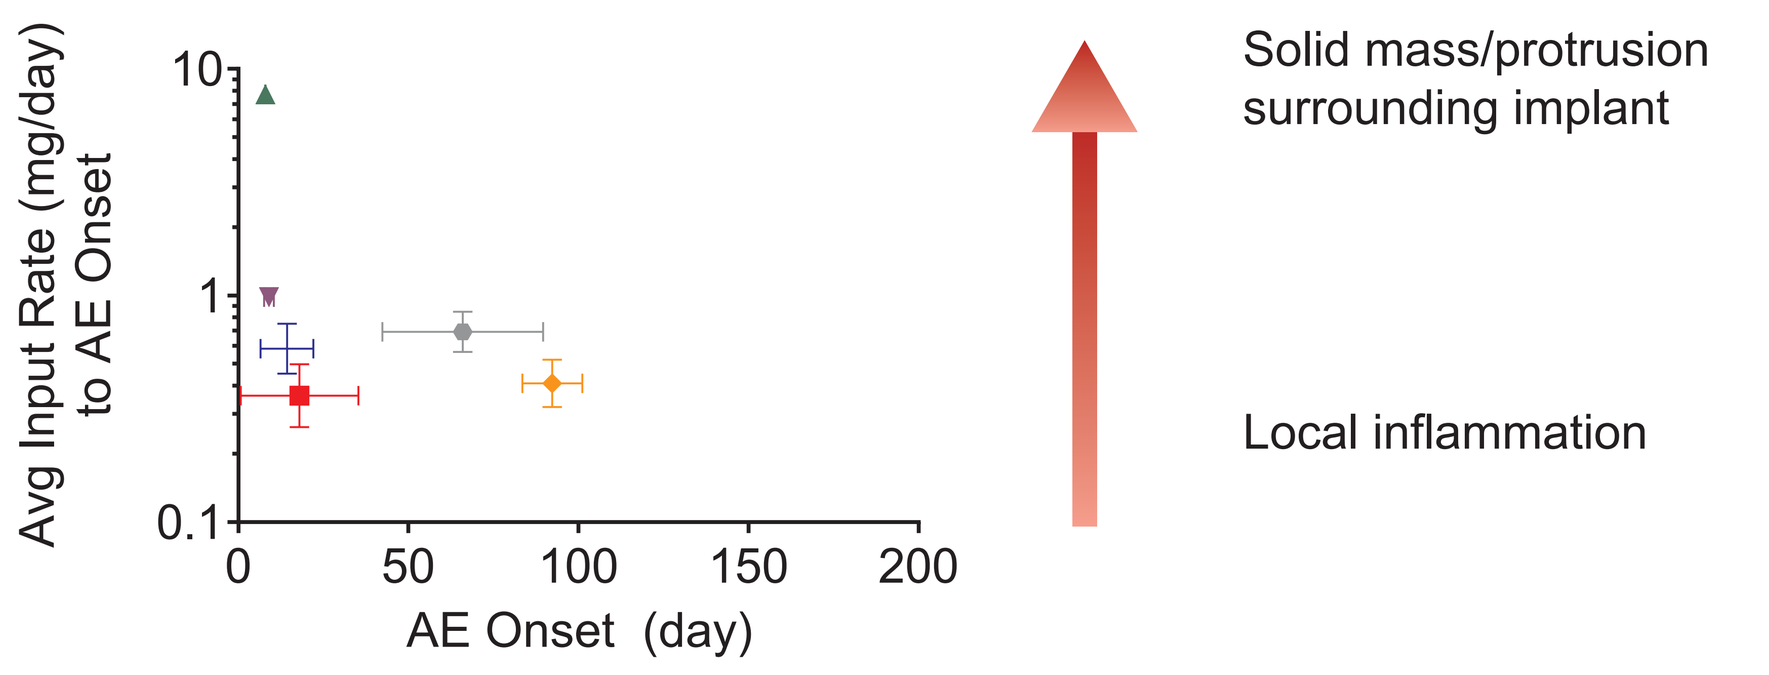
Avg Input Rate (mg/day) to AE Onset
AE Onset (day)
Solid mass/protrusion surrounding implant
Local inflammation

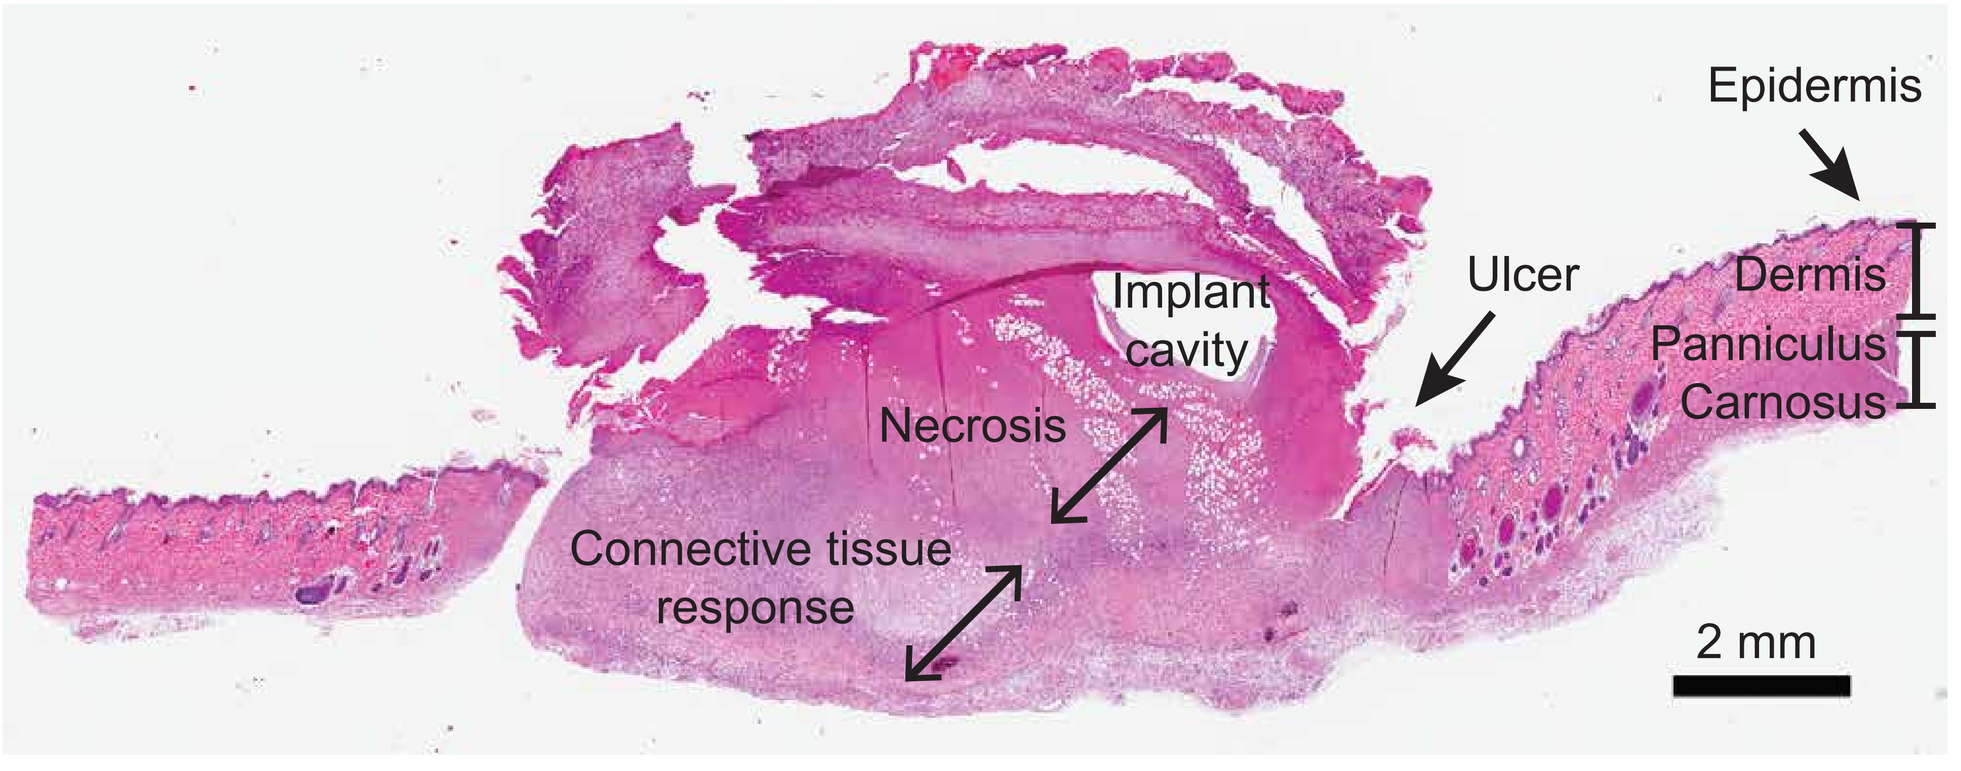
Epidermis
Ulcer
Dermis
Implant cavity
Panniculus Carnosus
Necrosis
Connective tissue response
2 mm

Hematoxylin- and Eosin-stained tissue around PU-coated tablet. Locally extensive necrosis proximal to the implant cavity is shown, along with a connective tissue response in the subdermal layers. The severity of the adverse event was proportional to the average ETV input rate of the implant.

Liver Entecavir-Triphosphate (ETV-TP) Detected at Day 142 or Longer in All Groups

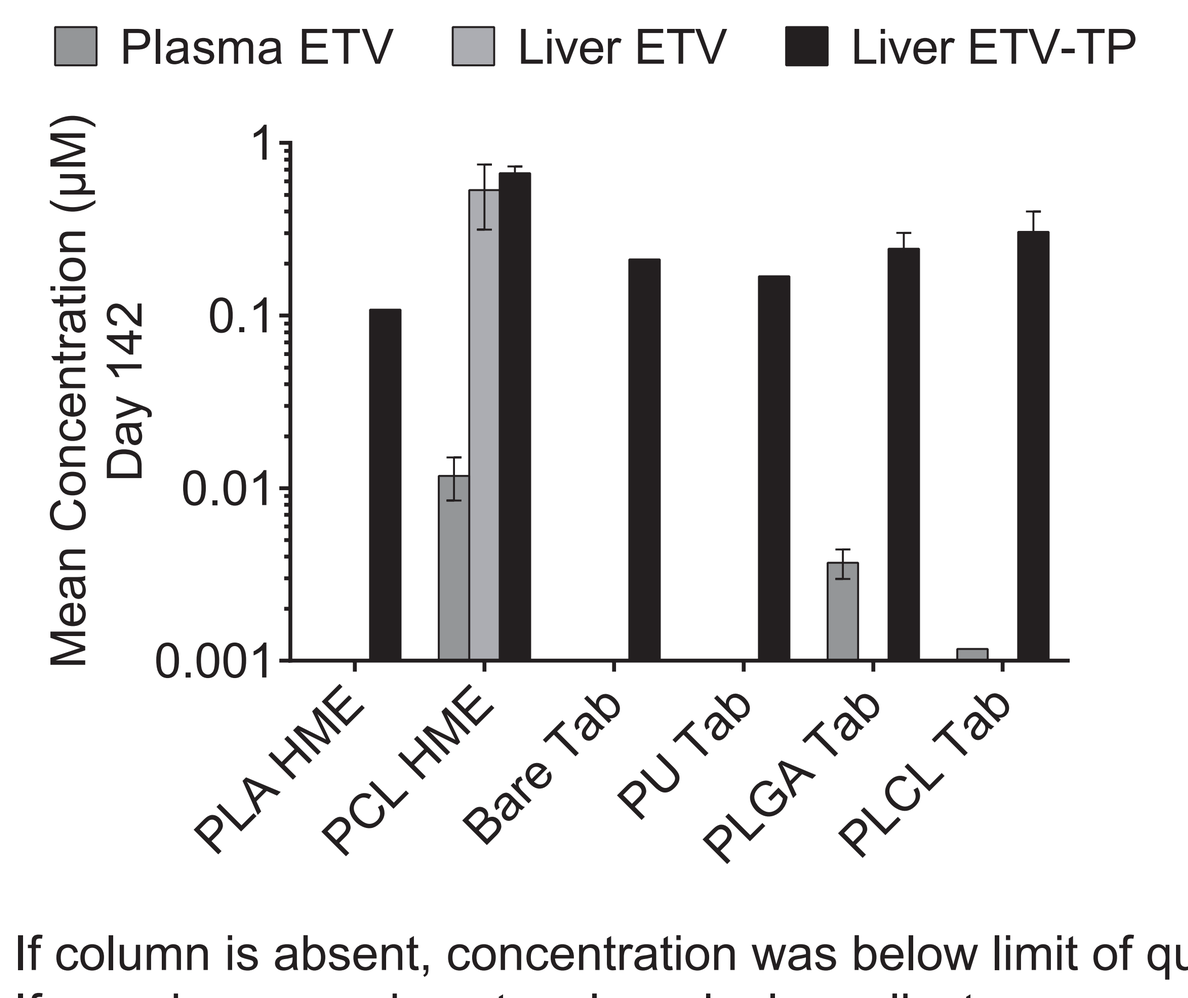
Plasma ETV
Liver ETV
Liver ETV-TP
Mean Concentration (µM) Day 142
PLA HME
PCL HME
Bare Tab
PU Tab
PLGA Tab
PLCL Tab

Assay Sensitivity:
$[ETV]_{plasma}$ >0.002µM
$[ETV]_{liver}$ >0.25µM
$[ETV\text{-}TP]_{liver}$ >0.1µM

If column is absent, concentration was below limit of quantitation at Day 142.
If error bars are absent, only a single replicate was measured at Day 142.

XRCT of Recovered Implants Reveals That Tissue Adhesion Alters HME Erosion Pathway

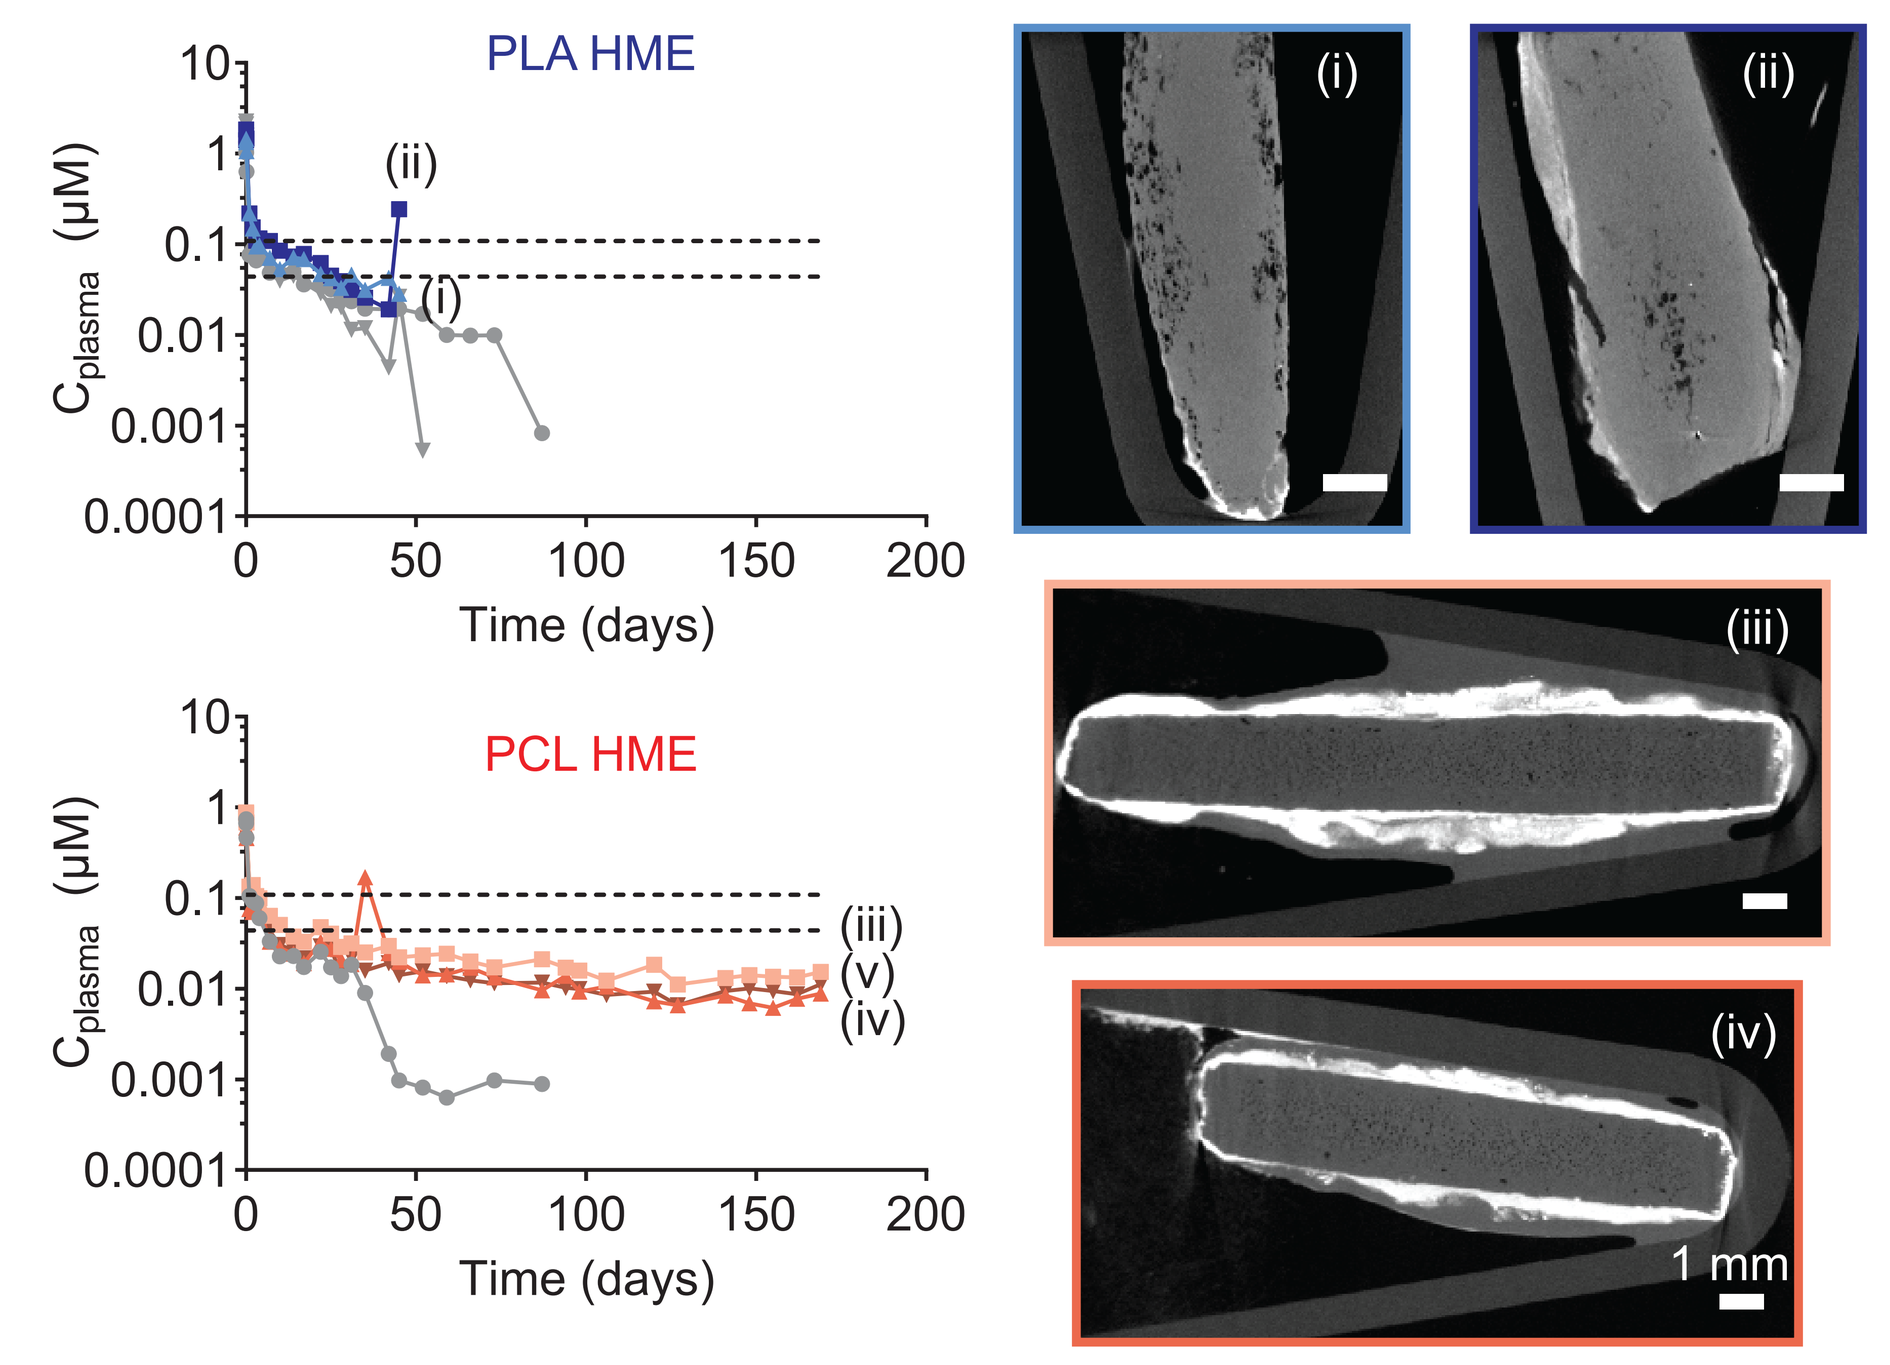
PLA HME
PCL HME
C_{plasma} (µM)
Time (days)
(i)
(ii)
(iii)
(iv)
(v)
1 mm

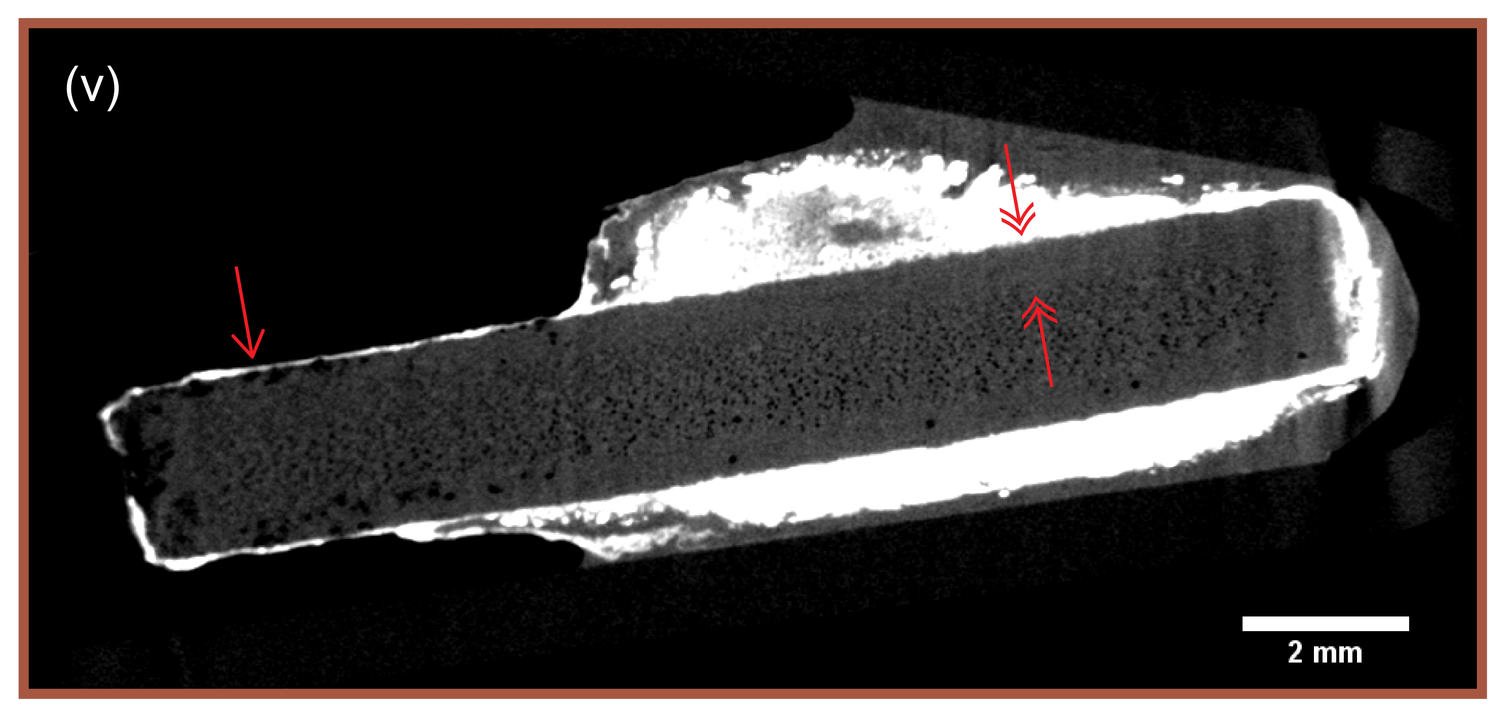
(v)
2 mm

PCL HME replicate (v) illustrates how extent of surface tissue adhesion correlates with erosion pathway. Single-headed arrow shows surface erosion in region with thin tissue adhesion. Double-headed arrows show core erosion in region with thick tissue.

Gamma Irradiation (25 kGy) Accelerates Release in Lactide-Containing Polymers

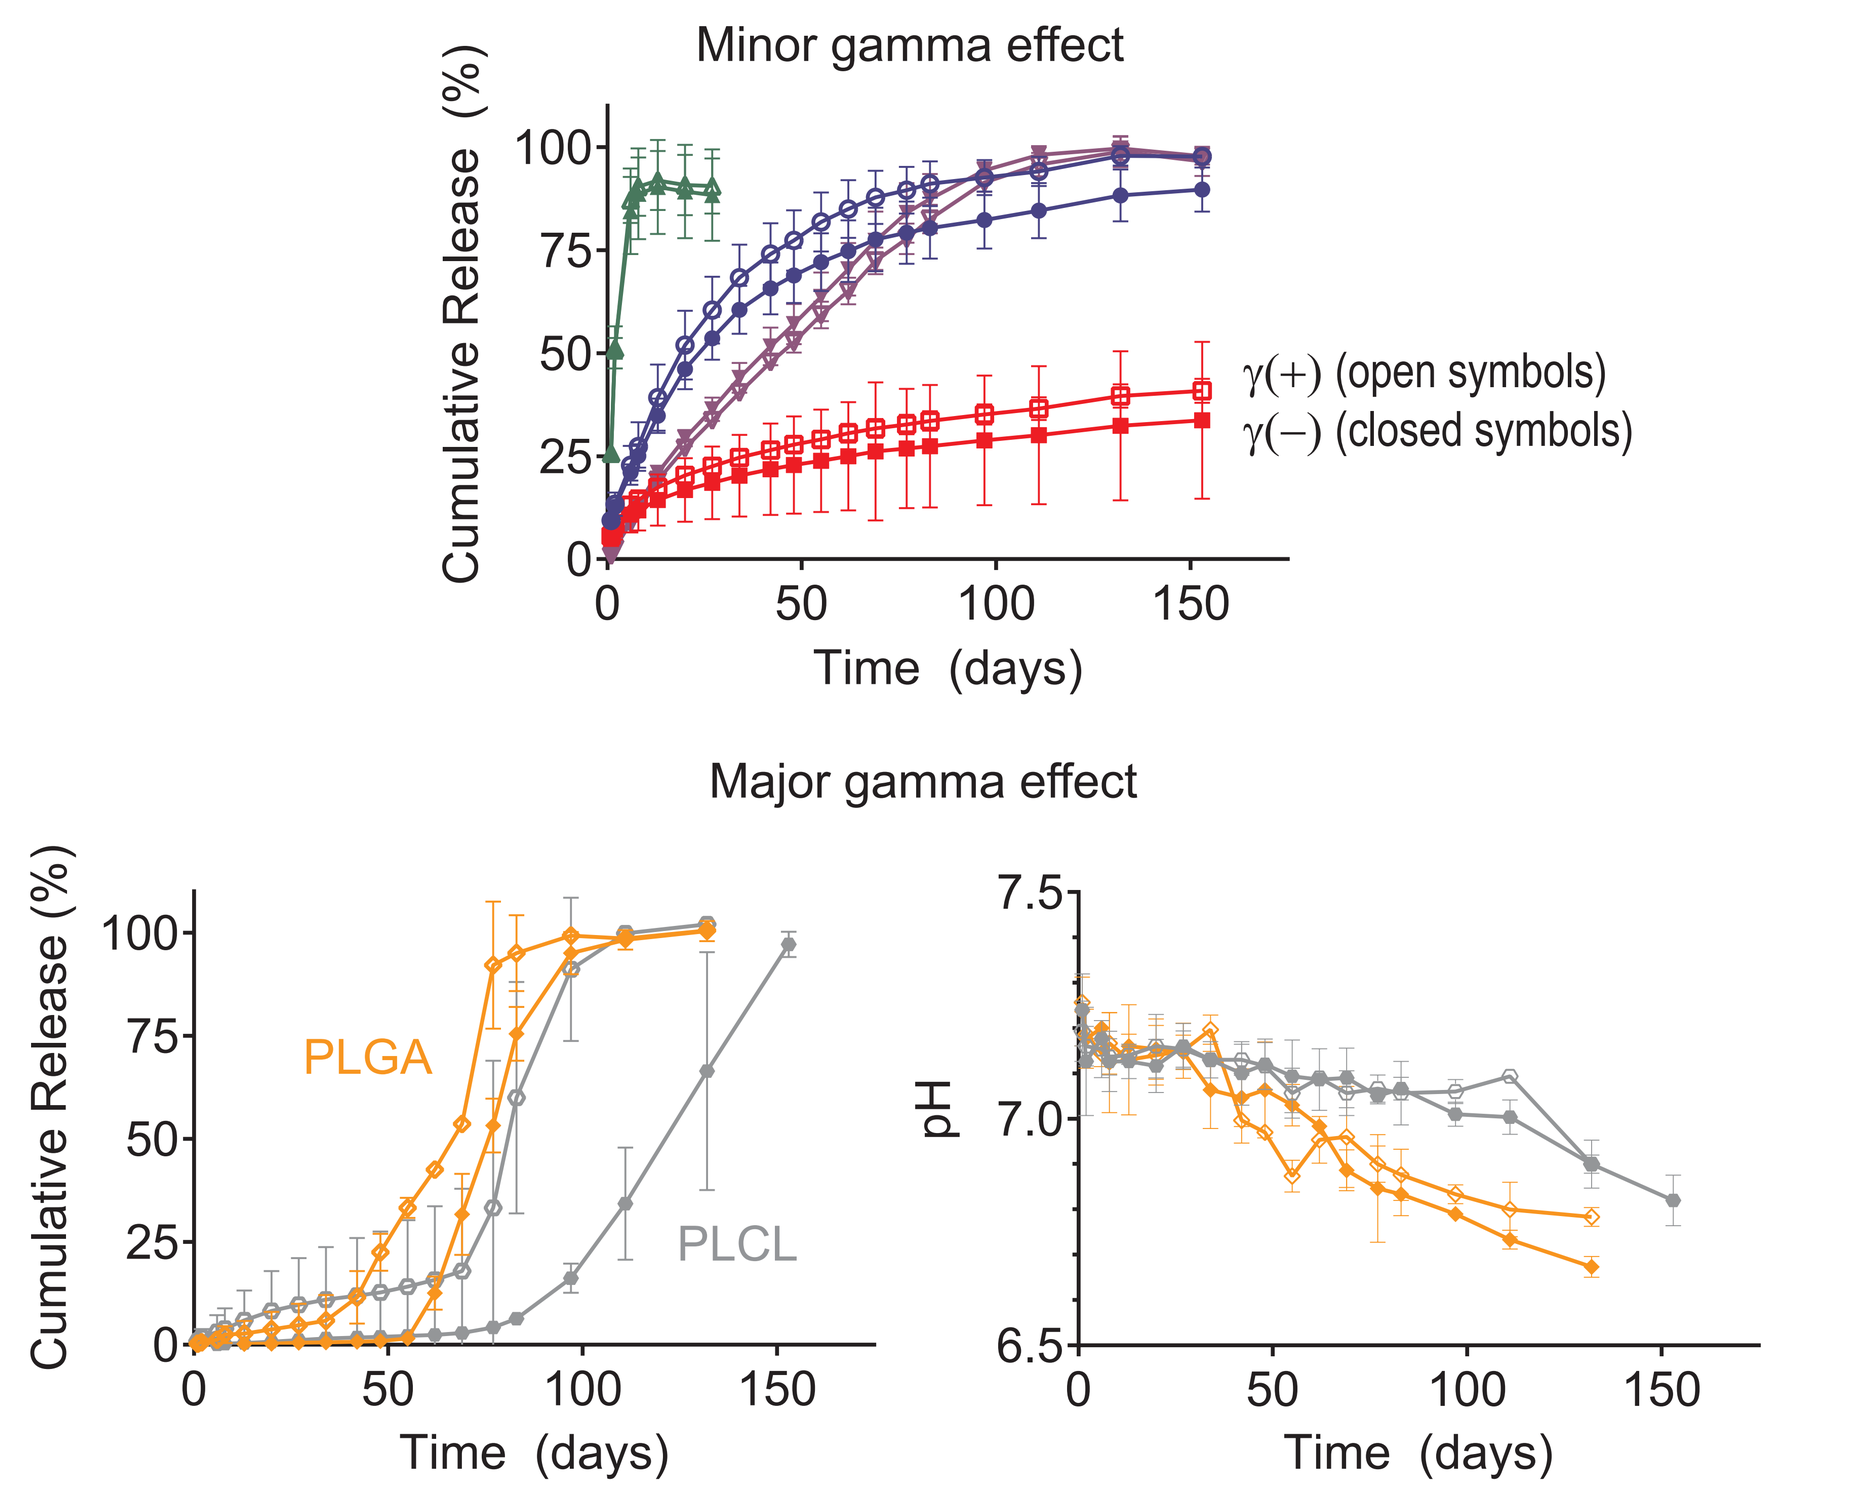
Minor gamma effect
Cumulative Release (%)
γ(+) (open symbols)
γ(−) (closed symbols)
Major gamma effect
PLGA
PLCL
pH
Time (days)

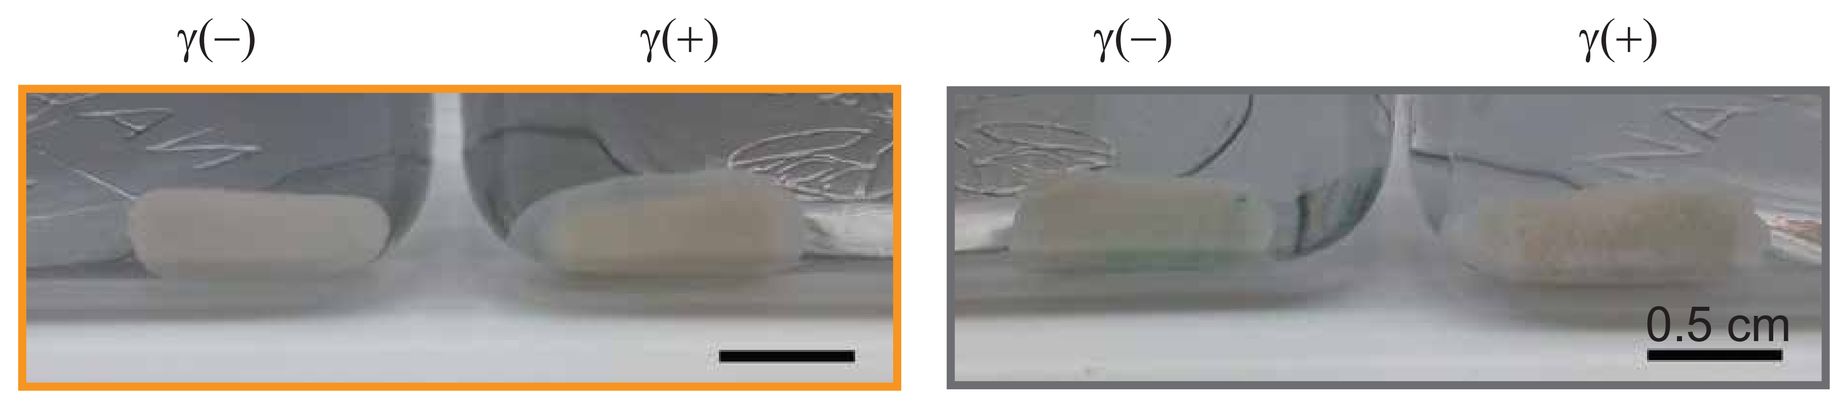
γ(−)
γ(+)
γ(−)
γ(+)
0.5 cm

PLGA Tab, Day 42 | PLCL Tab, Day 42

Conclusions

- Achieved an unprecedented 169 days of parenteral ETV release
- Of polymers tested, PCL HMEs provided most stable *in vivo* ETV blood plasma content
- Liver ETV-TP levels appear long-lived relative to plasma ETV. However, it is unclear if these ETV-TP levels are efficacious
- Local AEs at implantation site appear ETV specific, suggesting that polymer modalities may be applicable to better-tolerated drugs
- Differential erosion pathway of HMEs appears to correlate with tissue adhesion, but PK profiles were not differentiated
- Gamma irradiation accelerated *in vitro* release of lactide-containing polymers

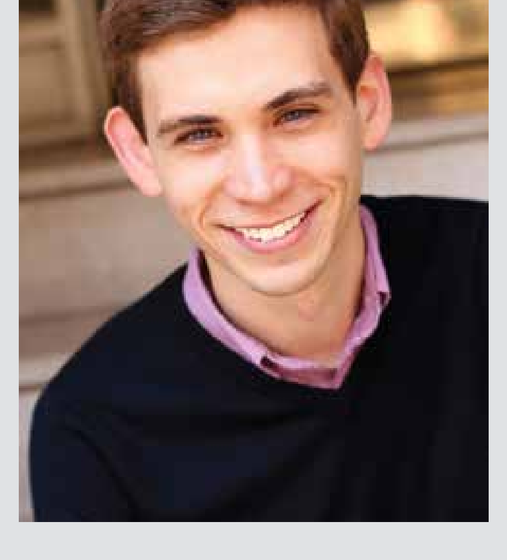

Steven J. Henry, PhD

Copyright © 2016 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All rights reserved.